Spatial transcriptomics reveals a transition from a prostate luminal to club-like cell state in 5-alpha reductase inhibitor treated BPH patients

*Diya Binoy Joseph[1], Gervaise H Henry[1], Alicia Malewska[1], Jeffrey C Reese[2], Ryan J Mauck[1], Jeffrey Gahan[1], Ryan C Hutchinson[1], James L Mohler[3], Claus G Roehrborn[1], Douglas W Strand[1]
[1]Department of Urology, UT Southwestern Medical Center, Dallas, TX 75390, USA, [2]Southwest Transplant Alliance, Dallas, TX 75231, USA
[3]Roswell Park Comprehensive Cancer Center, Buffalo, NY 14263, USA, *Presenting author (diya.binoyjoseph@utsouthwestern.edu)

UTSouthwestern Medical Center

Introduction

Understanding how prostate luminal cells adapt and survive in a low androgen environment is central to the clinical treatment of benign and malignant disease. 5-alpha reductase inhibitors (5ARIs) block the conversion of testosterone to dihydrotestosterone (DHT) and are used to treat Benign Prostatic Hyperplasia (BPH). 5ARI treatment reduces tissue DHT levels and causes involution of prostate glands. However, the histologic response to 5ARIs is heterogeneous and lower urinary tract symptoms in treated BPH patients often persist or worsen, leading to surgical intervention. We examined transcriptional changes accompanying 5ARI treatment to determine the cellular mechanisms of treatment resistance.

Approach

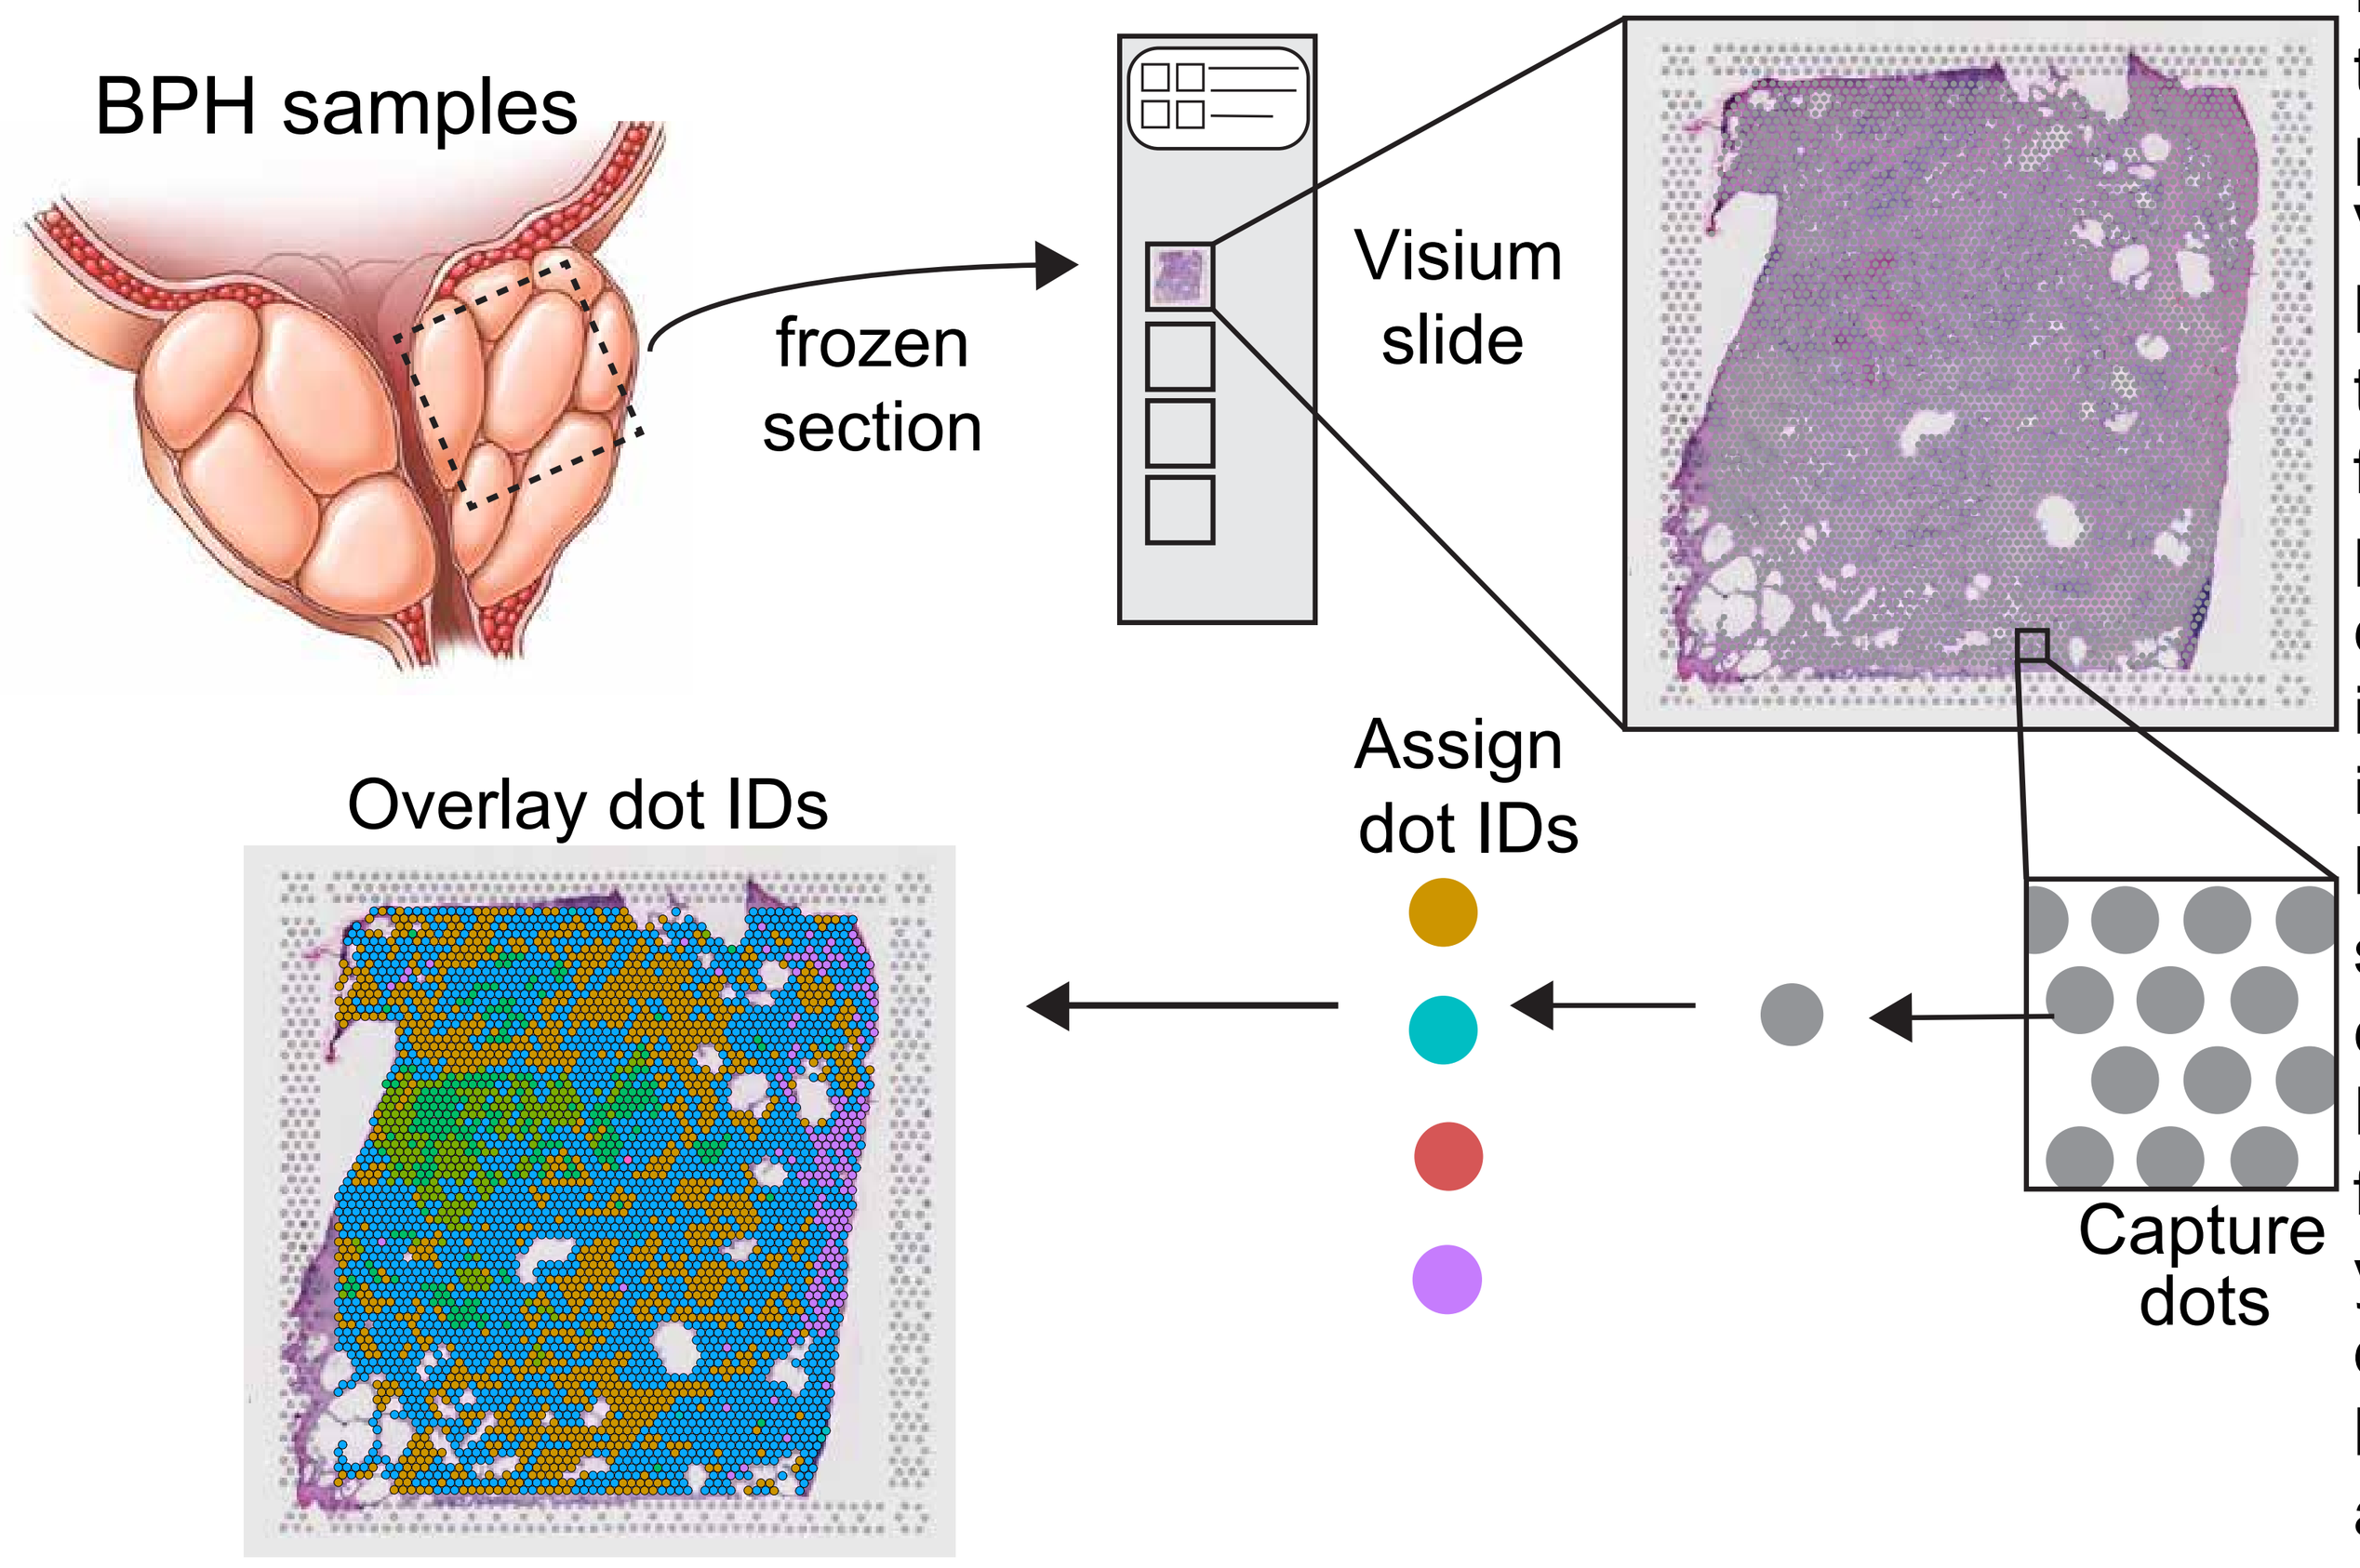

BPH samples
frozen section
Visium slide
Capture dots
Assign dot IDs
Overlay dot IDs

Frozen sections from untreated and 5ARI treated prostates were placed on Visium slides. The grid of barcoded capture dots on the slides binds to mRNA from the tissue. mRNA template is used to synthesise cDNA on the slide. Following the ligation of sequencing primers and indices, libraries are prepared and sequenced. Data is processed using Space Ranger from 10x genomics followed by secondary analysis using Seurat. Capture dots are assigned an ID based on their transcriptional similarity to our previous defined prostate cell types.

Atrophied regions have a club cell signature

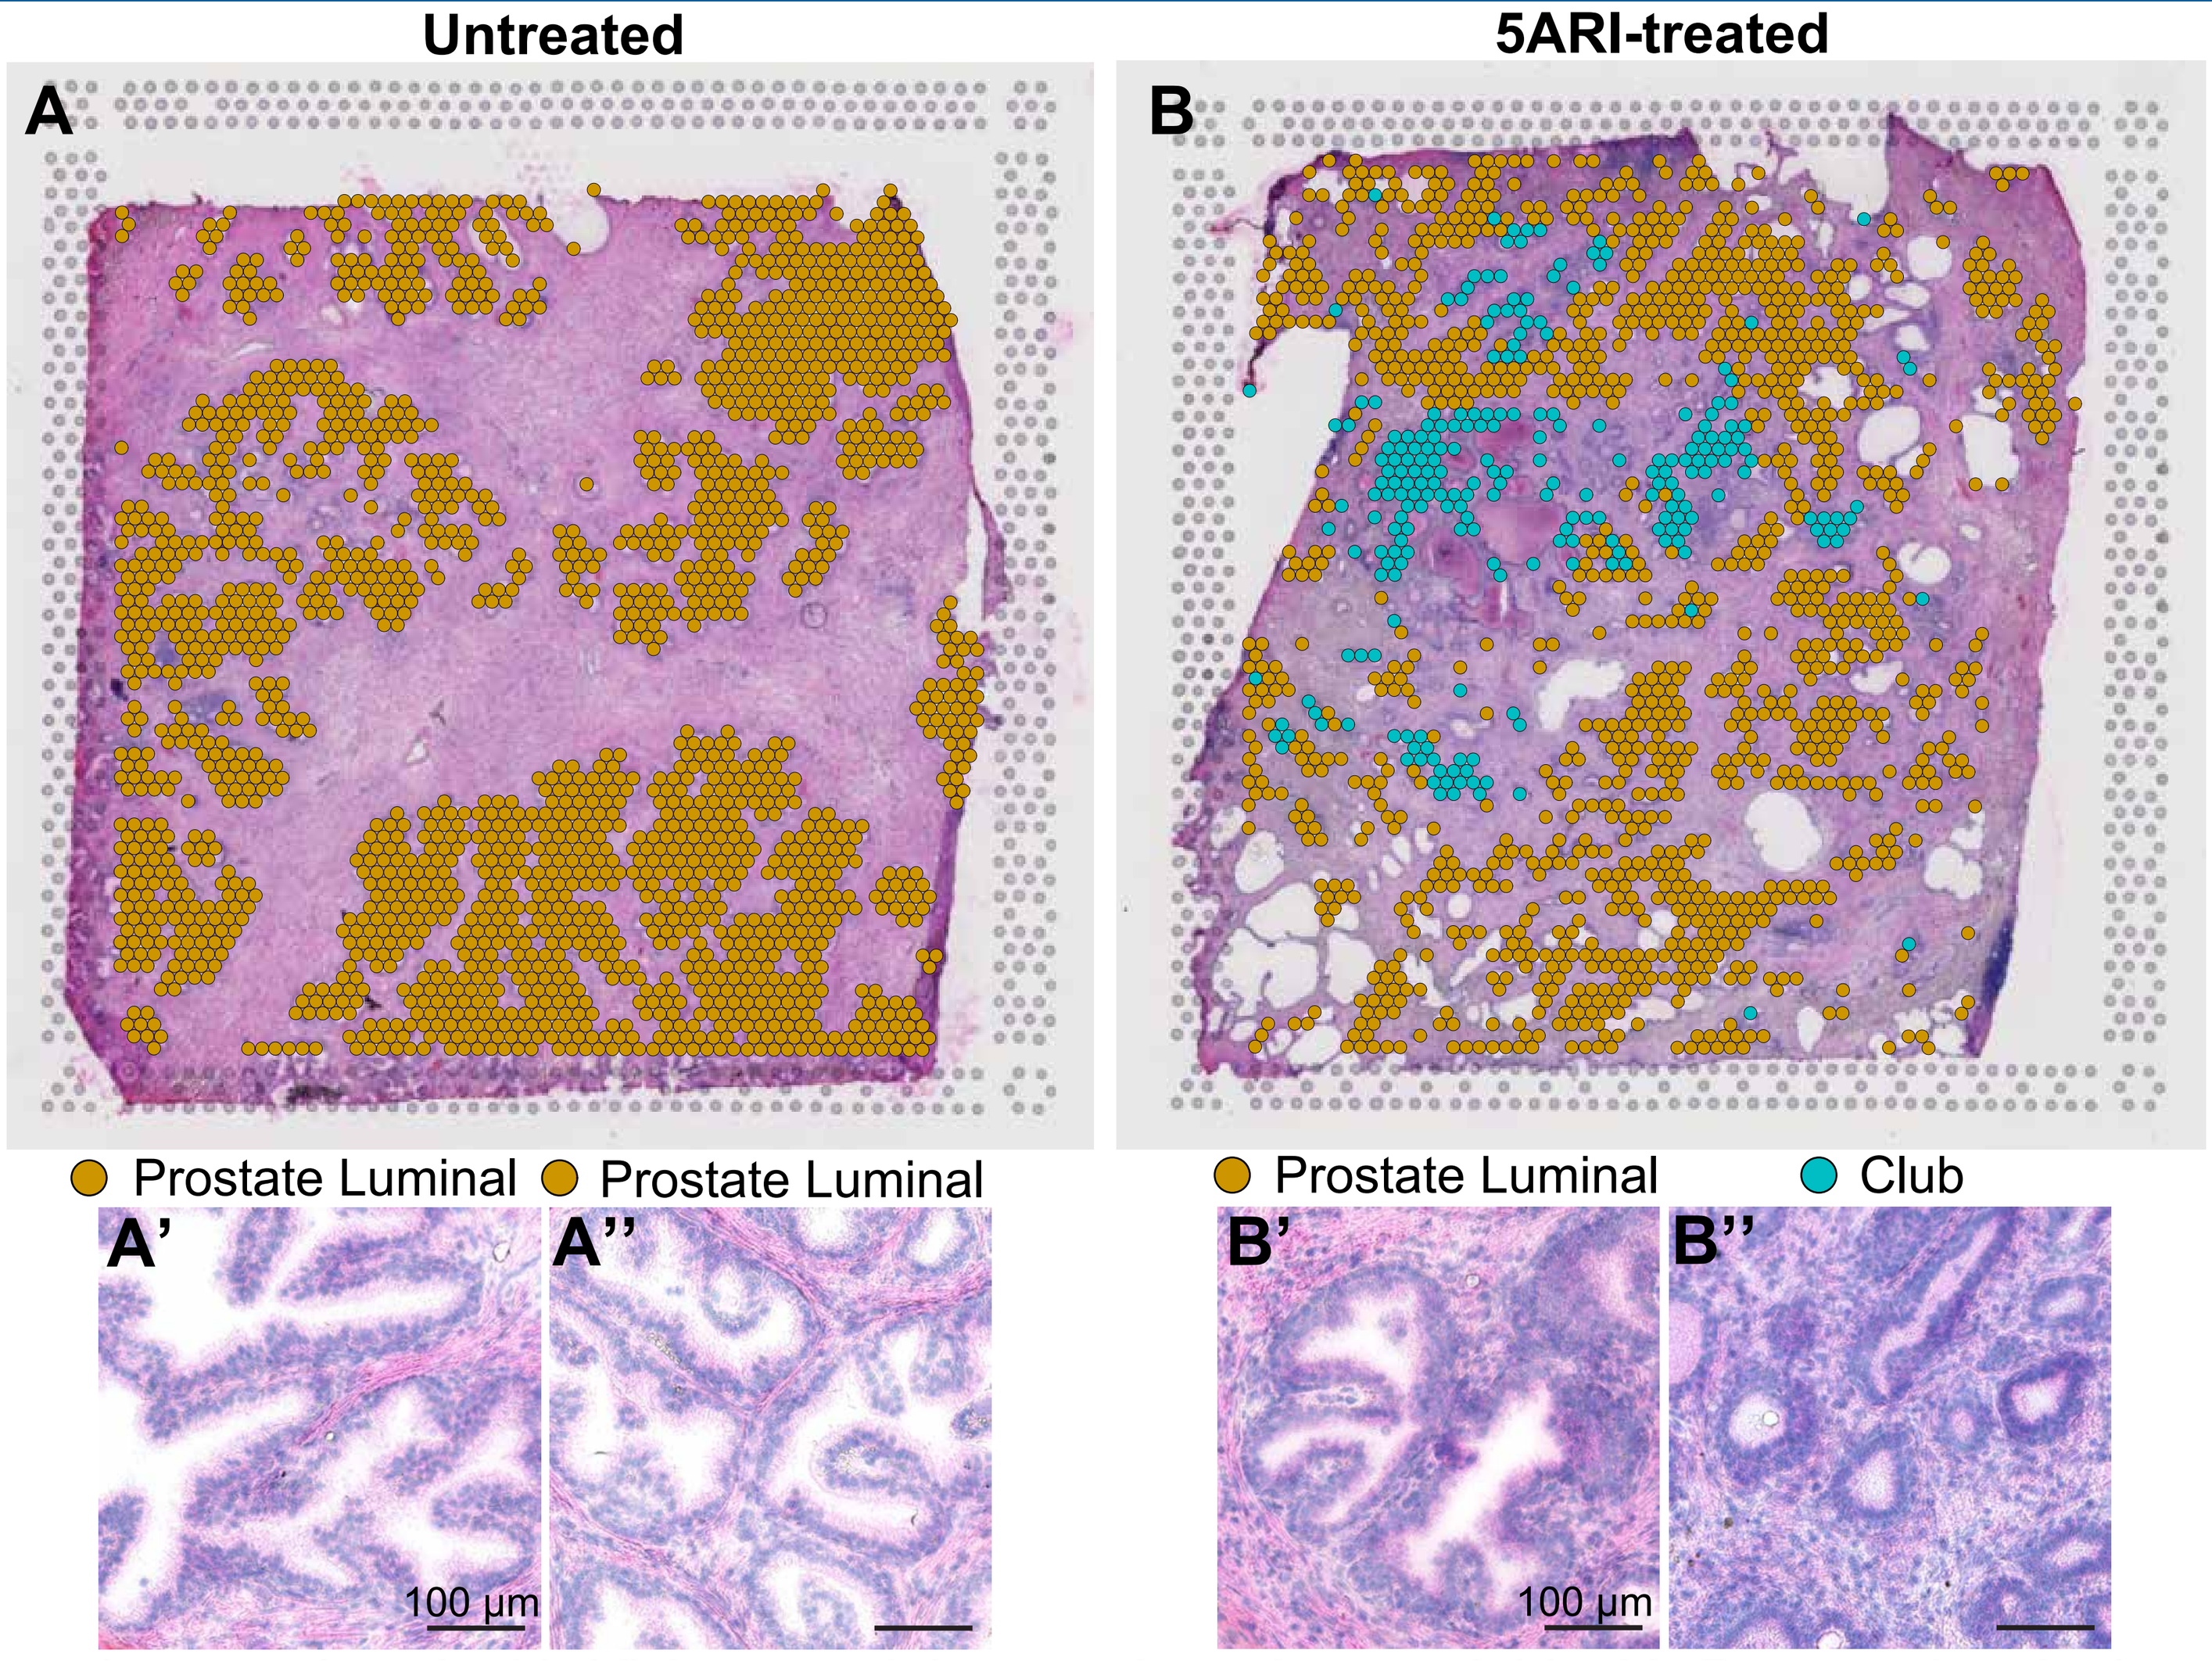

Untreated
5ARI-treated
A
B
Prostate Luminal
Prostate Luminal
Prostate Luminal
Club
A'
A''
B'
B''
100 µm
100 µm

Tissue sections analyzed with Visium spatial transcriptomics overlaid with Prostate Luminal and Club cell identities. Club cells are normally found in the urethra and transition zone in the normal prostate. Club cells are absent in untreated prostates (A) but their transcriptional signature is increased in regions of 5ARI atrophy (B).

Prostate luminal to club-like adaptation

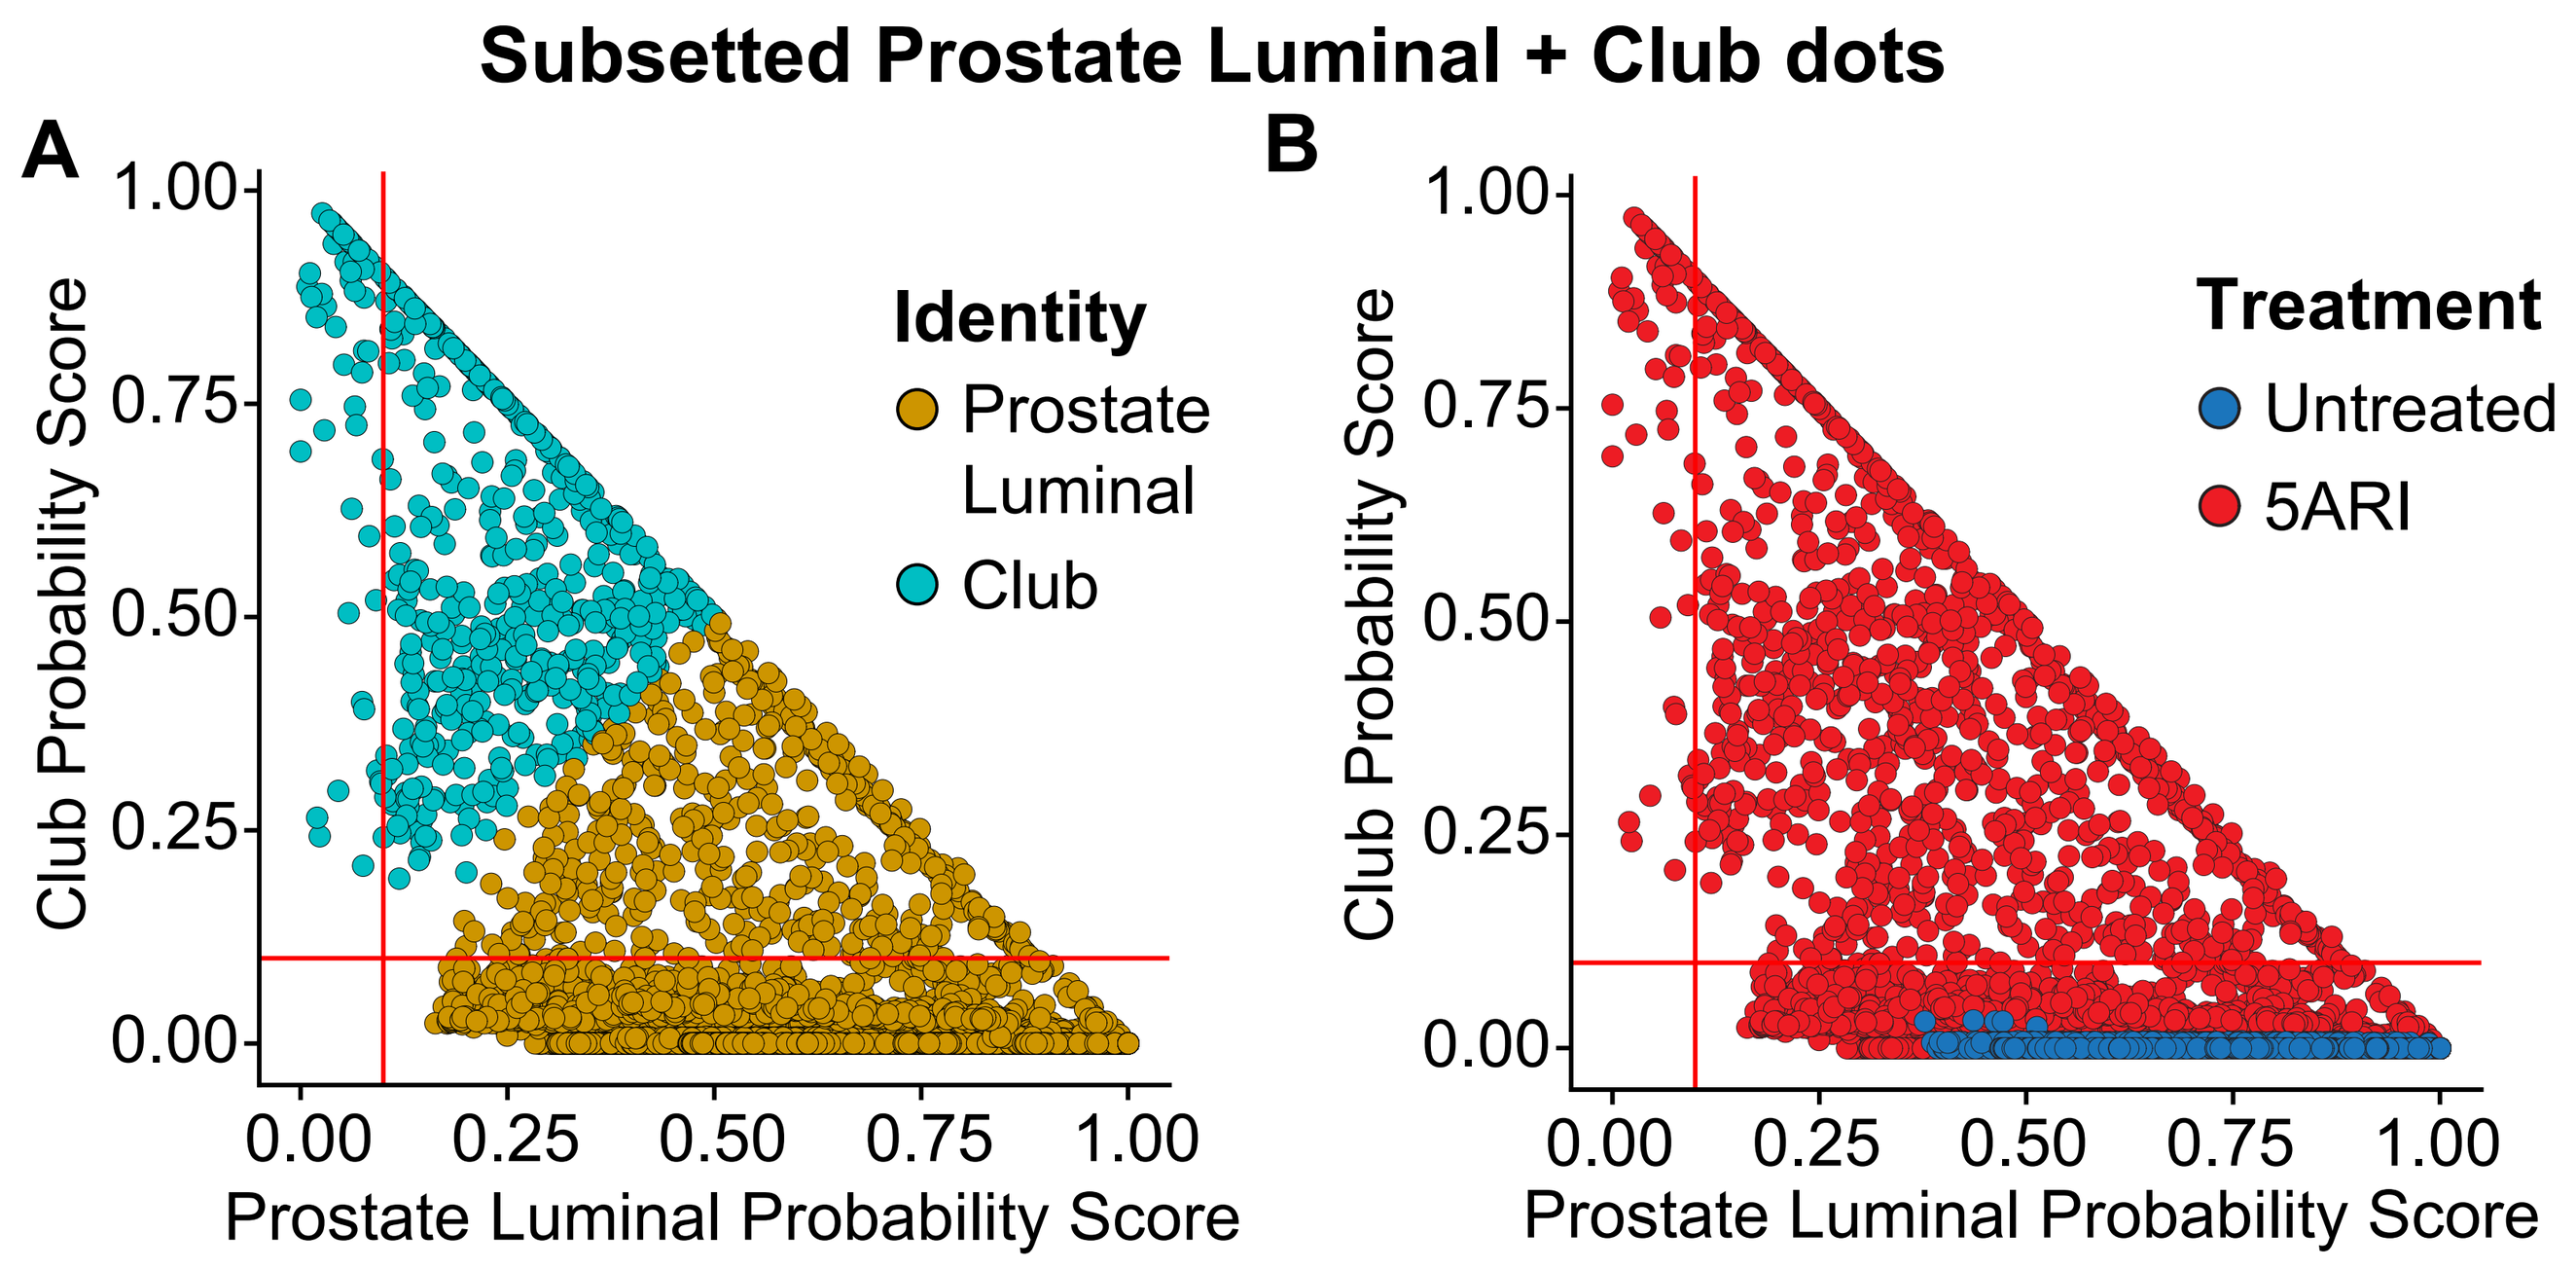

Subsetted Prostate Luminal + Club dots
A
B
Club Probability Score
Prostate Luminal Probability Score
0.00
0.25
0.50
0.75
1.00
Identity
Prostate Luminal
Club
Treatment
Untreated
5ARI

Prostate luminal epithelia undergo a transition to a club-like state in 5ARI treated prostates. Plotting dots that identify as prostate luminal and club against probability scores of being prostate luminal or club, we found that 5ARI treated samples displayed a prostate luminal to club spectrum (A). Notably, untreated samples were largely prostate luminal cells with a low club probability score (B).

Morphological changes in club-like adaptation

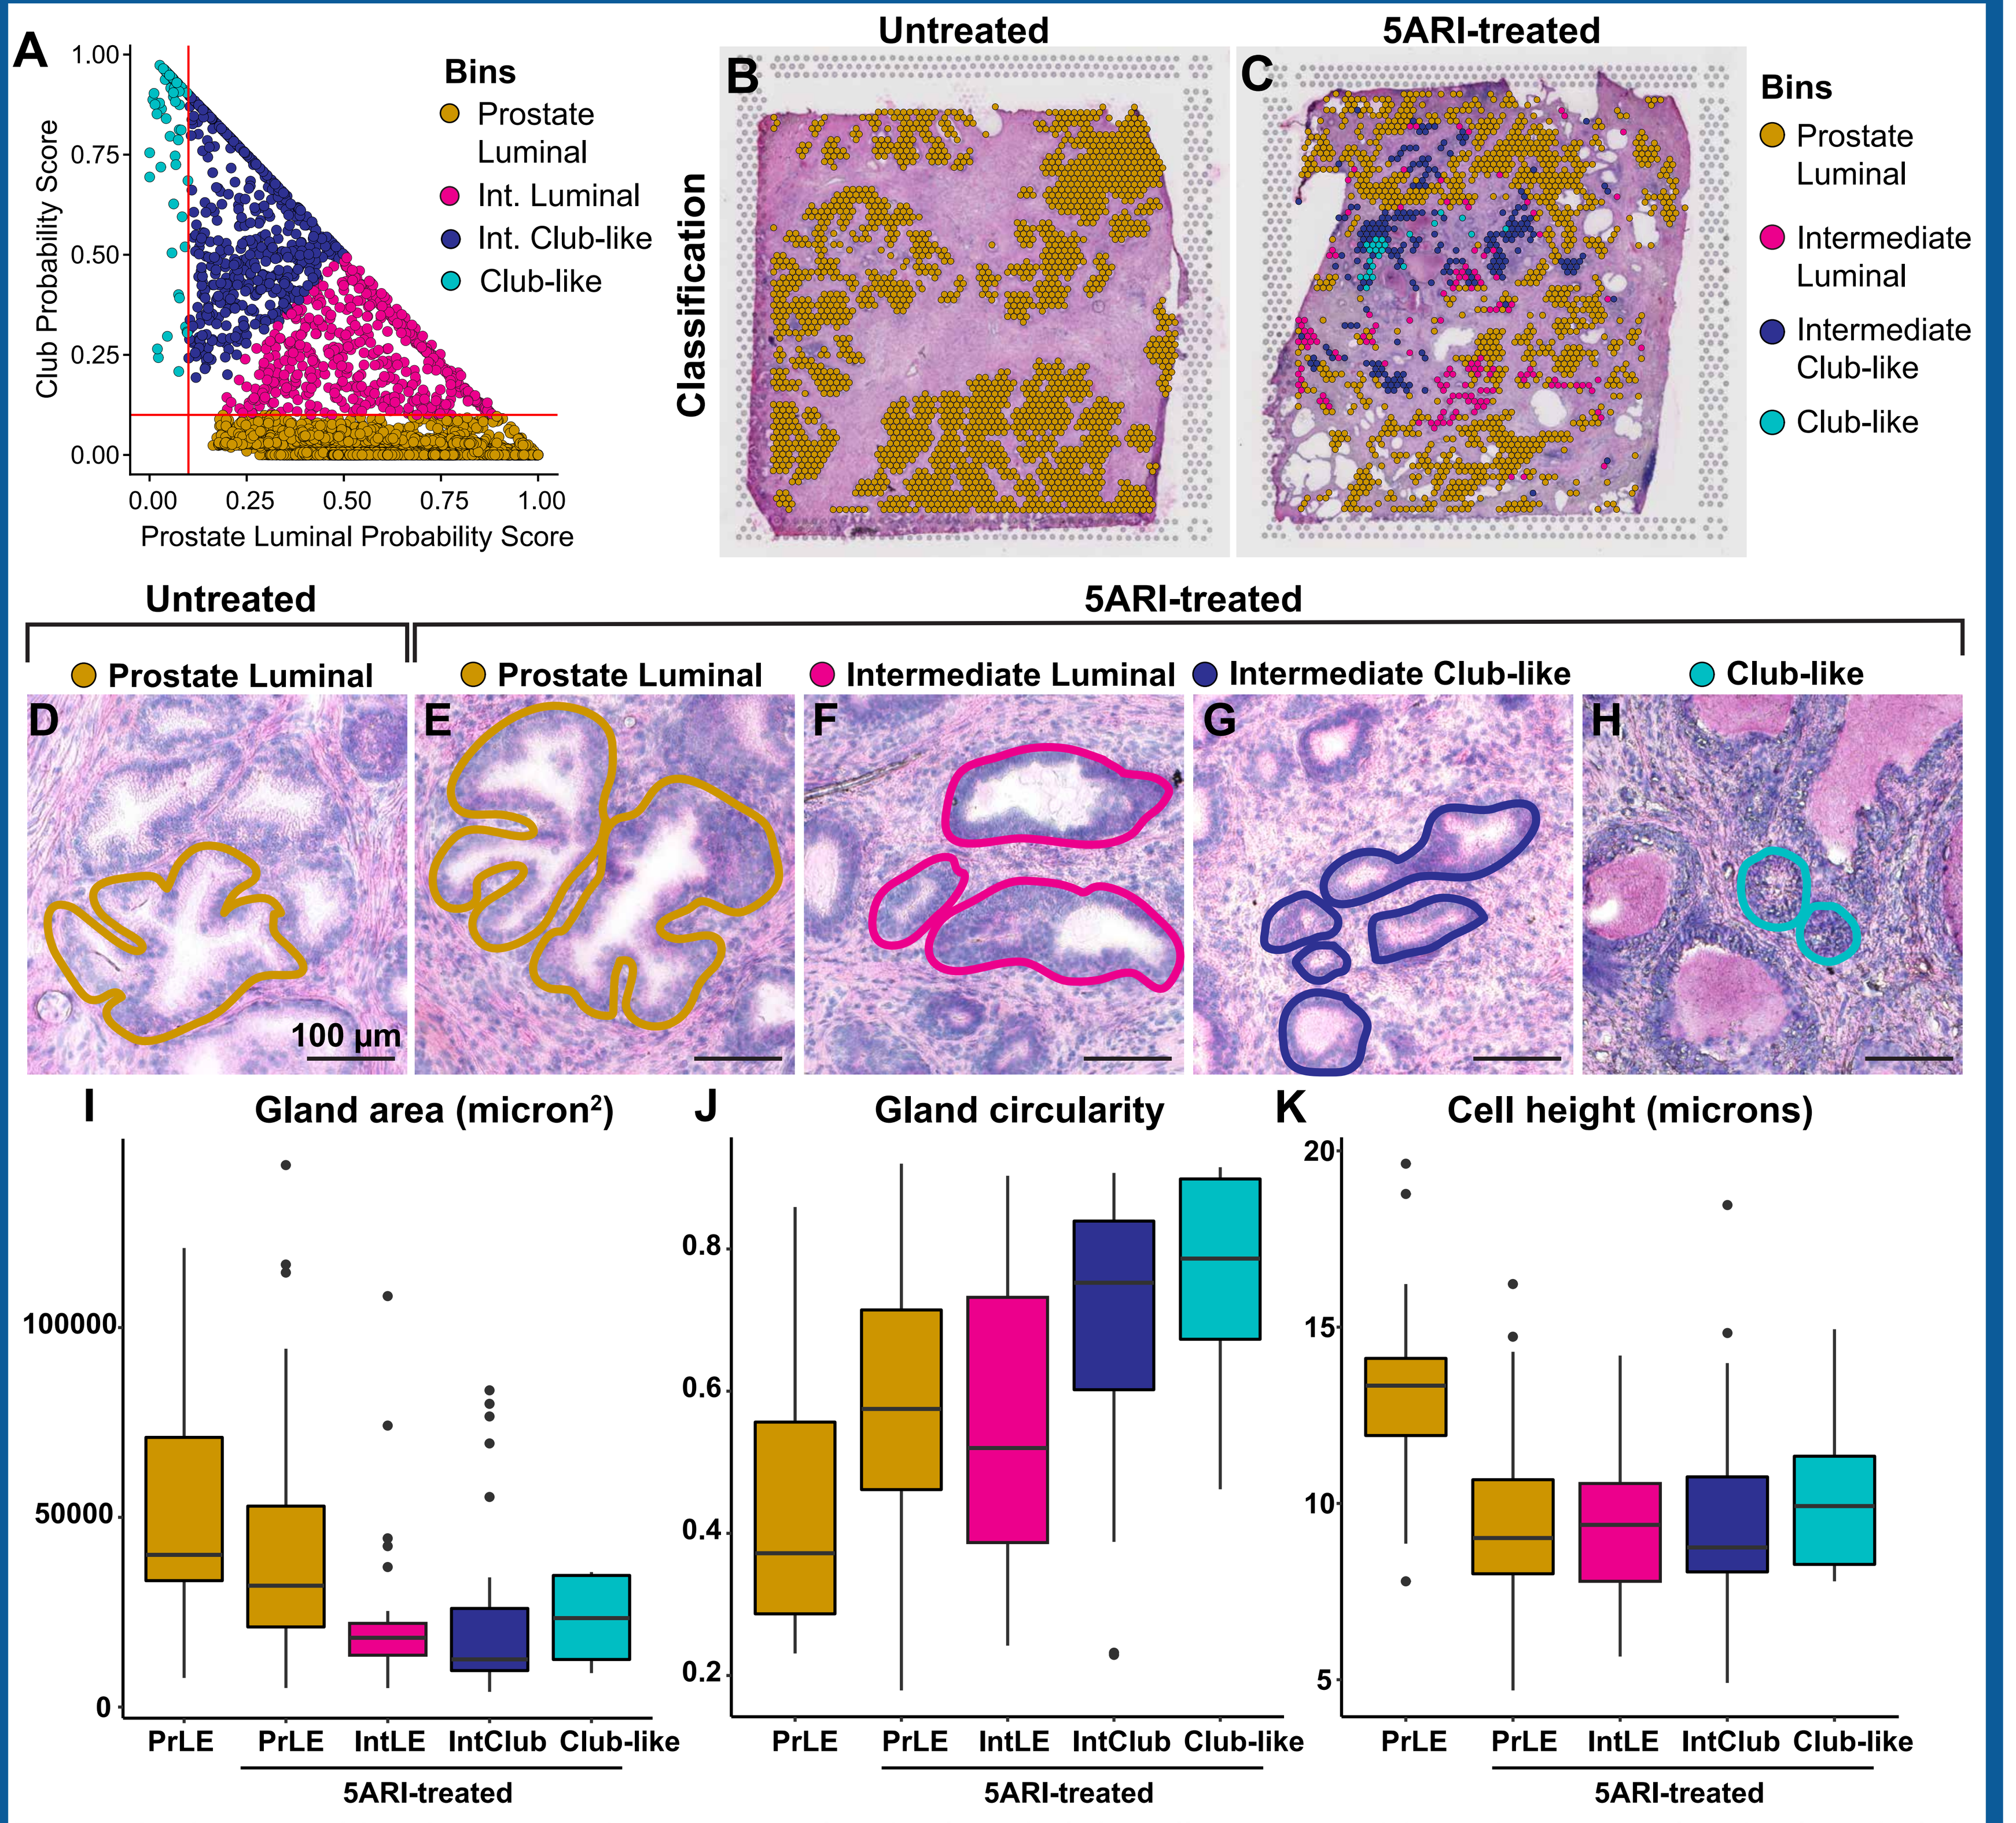

A
Club Probability Score
Prostate Luminal Probability Score
Bins
Prostate Luminal
Int. Luminal
Int. Club-like
Club-like
Untreated
5ARI-treated
B
C
Classification
Bins
Prostate Luminal
Intermediate Luminal
Intermediate Club-like
Club-like
Untreated
5ARI-treated
Prostate Luminal
Prostate Luminal
Intermediate Luminal
Intermediate Club-like
Club-like
D
E
F
G
H
100 µm
I
Gland area (micron²)
J
Gland circularity
K
Cell height (microns)
PrLE
PrLE
IntLE
IntClub
Club-like
5ARI-treated

To study the transition between the prostate luminal and club cell state, we binned prostate luminal and club dots into 4 categories (A). The categories were overlaid on the tissue sections on which the Visium experiment had been performed (B-C). The glands underlying the different categories were manually outlined in ImageJ (D-H). Using in built functions in ImageJ, gland area (I) and gland circularity (J) were calculated. Gland area decreased after 5ARI treatment compared to untreated. Gland area decreased as glands transitioned from a luminal to a club-like phenotype. Similarly, circularity increased in 5ARI treated glands compared to untreated. Gland circularity was highest in club-like categories indicating a loss of epithelial infoldings. Prostate luminal cell heights (K) decreased after 5ARI treatment compared to the untreated condition.

Decrease in AR signaling and increase in club genes

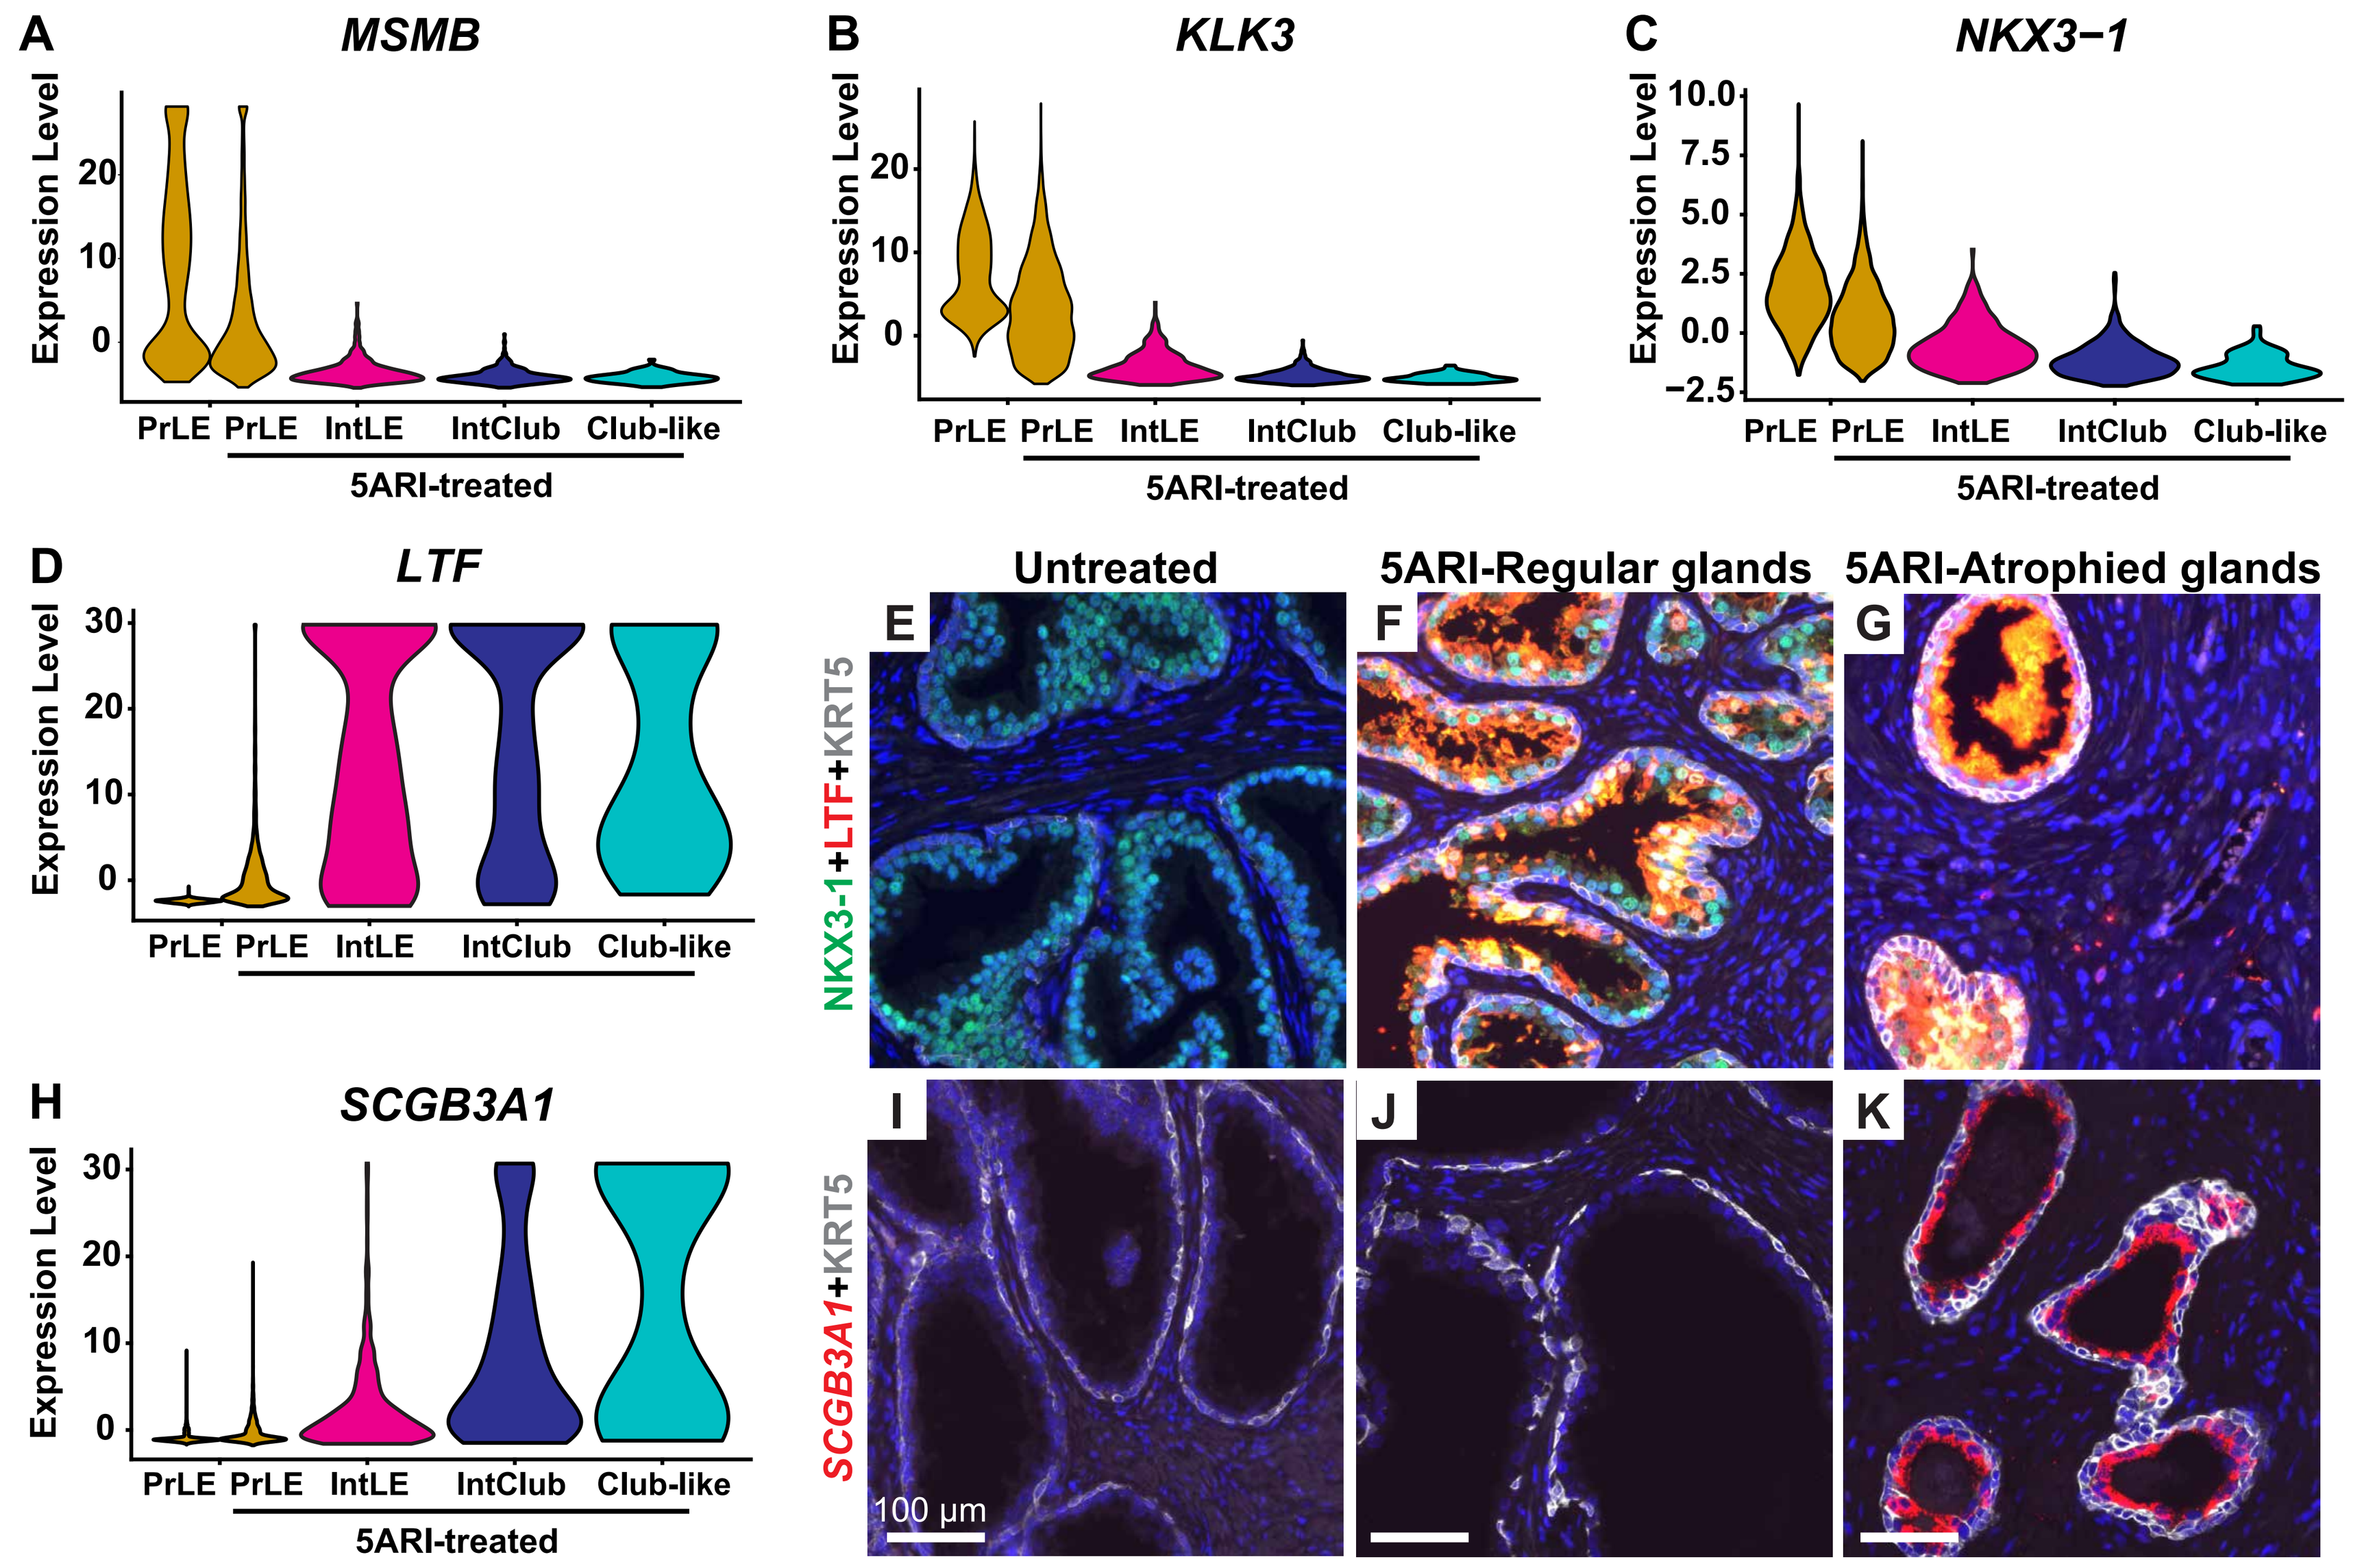

A
MSMB
B
KLK3
C
NKX3−1
D
LTF
H
SCGB3A1
Expression Level
PrLE
PrLE
IntLE
IntClub
Club-like
5ARI-treated
Untreated
5ARI-Regular glands
5ARI-Atrophied glands
E
F
G
I
J
K
NKX3-1+LTF+KRT5
SCGB3A1+KRT5
100 µm

Expression of androgen regulated genes *MSMB* (A), *KLK3/PSA* (B) and the prostate specific transcription factor *NKX3-1* (C) decreased after treatment with 5ARIs. A reciprocal increase in the club cell markers LTF (D-G) and SCGB3A1 (H-K) was observed in 5ARI treated prostates compared to untreated prostates. Specifically, regions of atrophy displayed increased expression of club cell markers.

Increase in NF-κB activation in atrophied regions

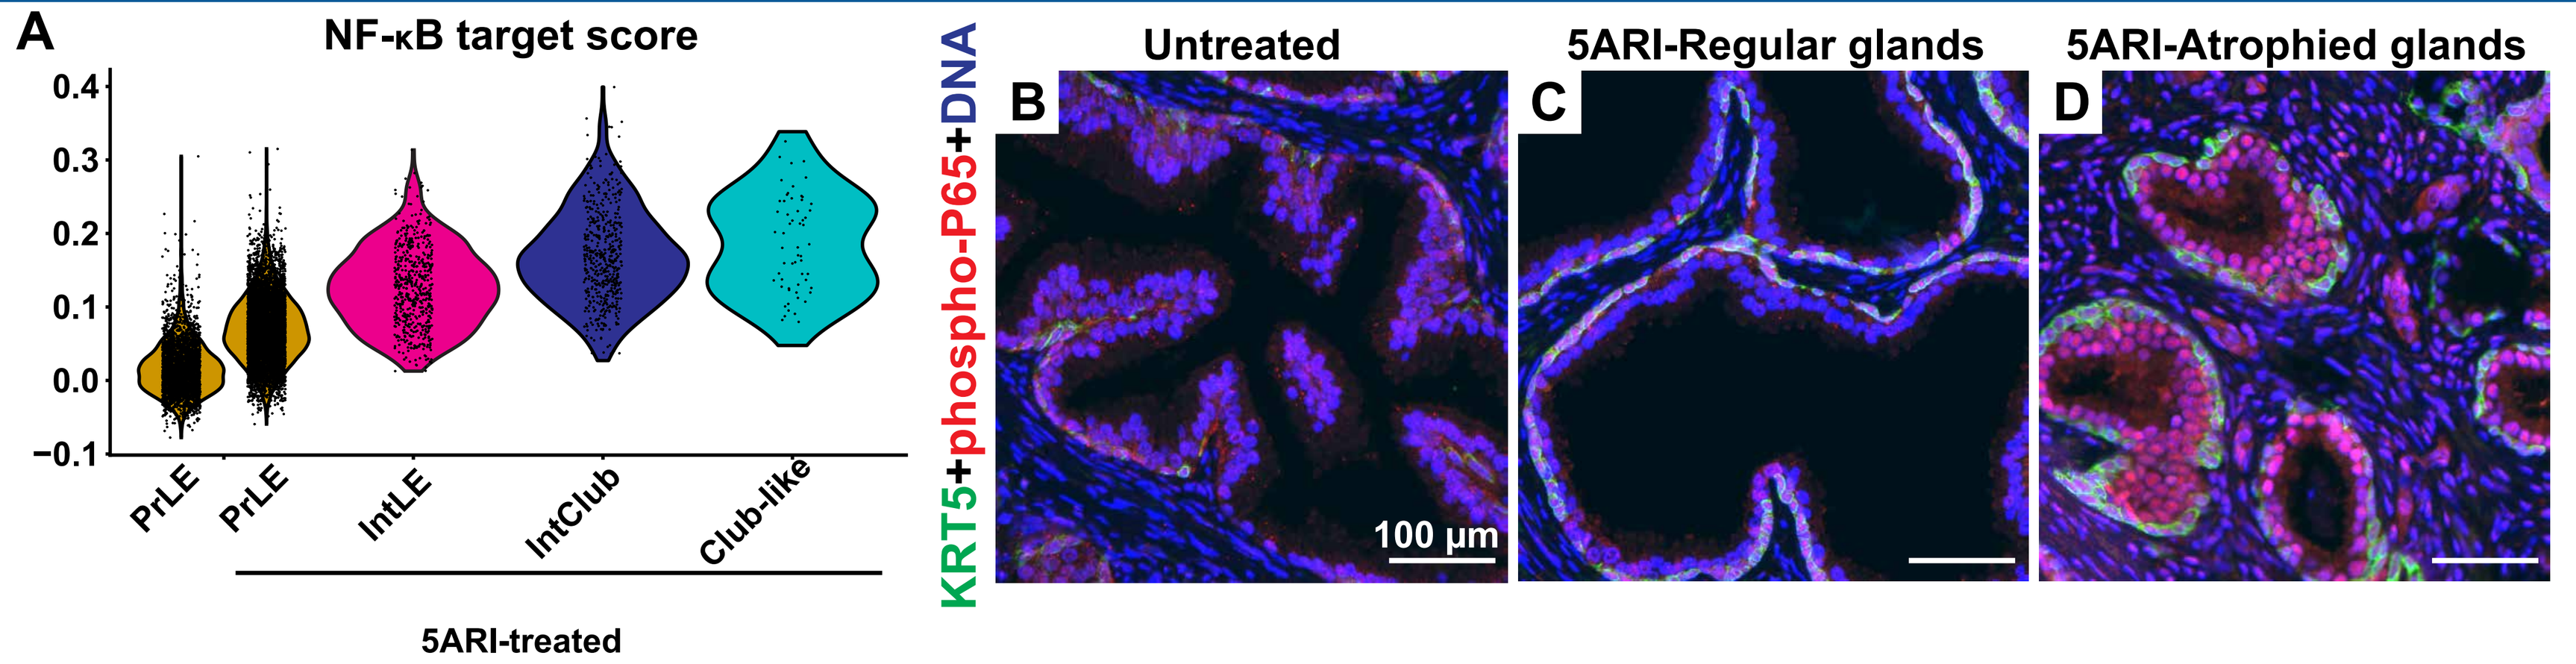

A
NF-κB target score
PrLE
PrLE
IntLE
IntClub
Club-like
5ARI-treated
KRT5+phospho-P65+DNA
Untreated
5ARI-Regular glands
5ARI-Atrophied glands
B
C
D
100 µm

A custom list of NF-κB target genes from *Pahl (1998), Oncogene* was used to compute a target score which showed increased NF-κB activation after 5ARI treatment (A). Nuclear localization of activated phospho-p65/RELA, a key NF-κB subunit was increased in regions of 5ARI induced atrophy (B-D).

Conclusions

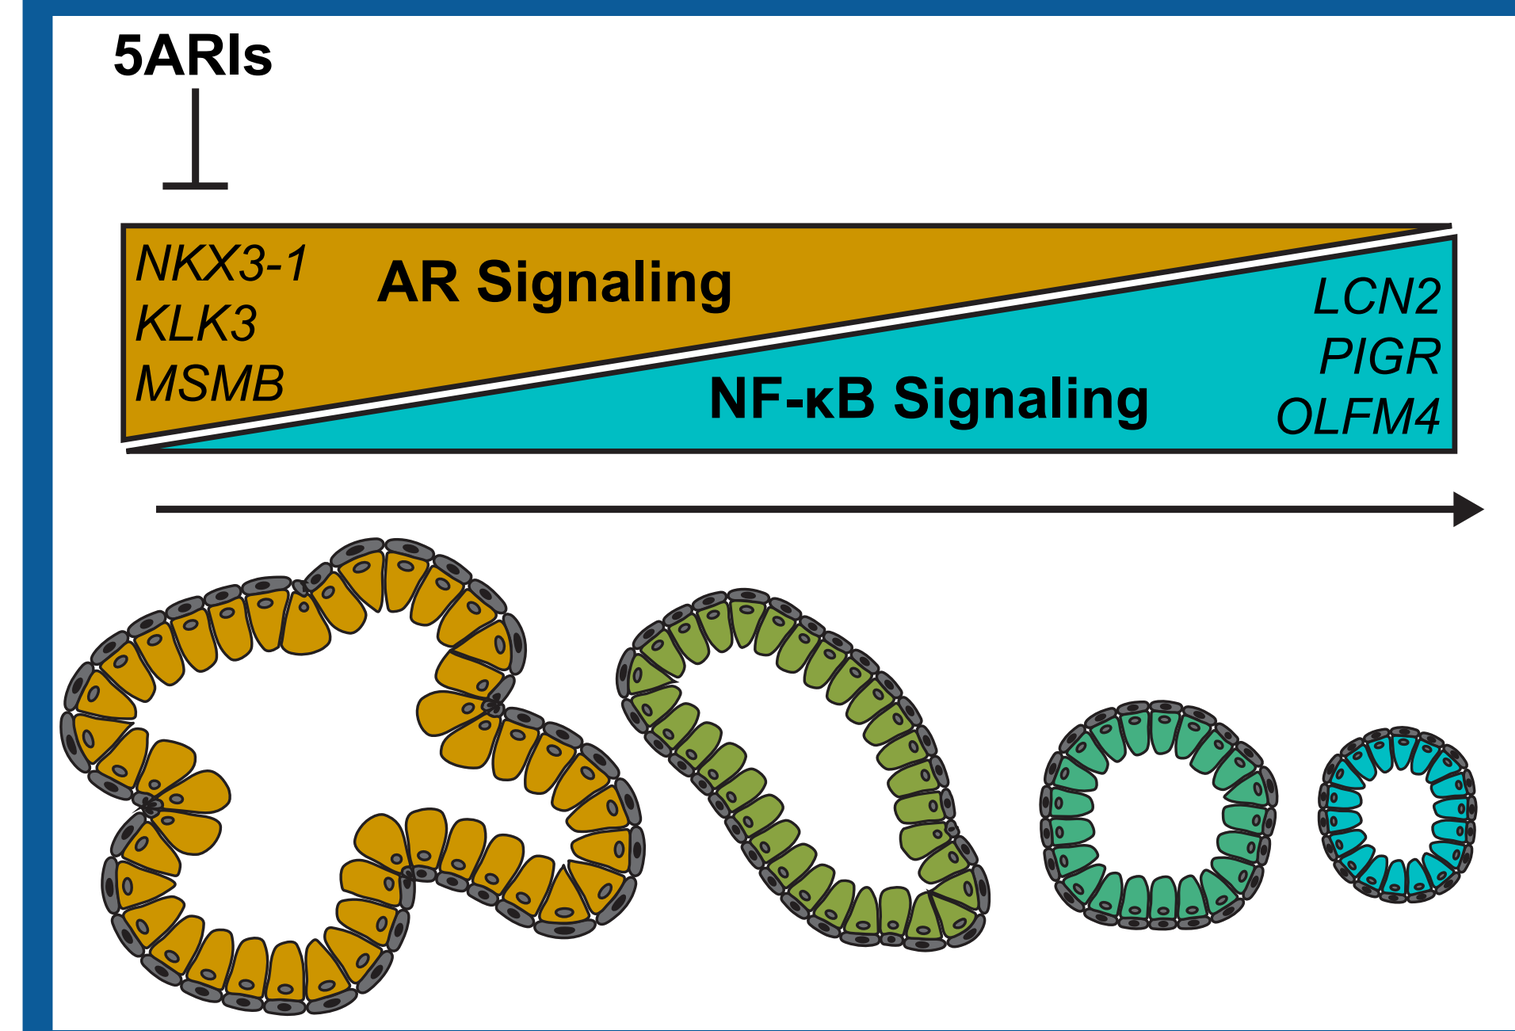

5ARIs
NKX3-1
KLK3
MSMB
AR Signaling
NF-κB Signaling
LCN2
PIGR
OLFM4

1. 5ARI response is heterogeneous and results in localized atrophy.

2. Atrophied regions display a club-like signature and have loss of androgen receptor signaling.

3. Atrophied regions have increased NF-κB activation.

4. Epithelial cells in prostate glands undergo a survival adaptation from a prostate luminal to a club-like state in response to low androgen levels.

Acknowledgements

Funding: AUA Research Scholar Award 659333 to DBJ, R01 DK115477 to DWS, NCI P30CA016056 to Roswell Park Comprehensive Cancer Center. We want to thank the staff and surgeons at the Southwest Transplant Alliance and UT Southwestern, as well as the tissue donors and their families for their commitment to basic science research.